Intravitreal Delivery of Ixo-vec for Neovascular Age-Related Macular Degeneration Results in Widespread Expression of Aflibercept in Cynomolgus Monkey Eyes as Confirmed Using In-Situ Hybridization.

Julio Delano Nieves[1], Gustavo de Alencastro[1], Kelly Michiko Hanna[1], Jenny Vo[1], Stephanie Lussier[1], Ruslan N. Grishanin[1], Brigit E. Riley[1];

[1]Adverum Biotechnologies Redwood City, CA

ADVERUM

BACKGROUND

- Ixo-vec is a novel gene therapy biofactory product engineered for wide-spread pan retinal expression of Aflibercept for the treatment of neovascular age-related macular degeneration
- Here we evaluated the nonclinical AAV per cell vector genome (vg) biodistribution and mRNA expression of intravitreal (IVT)-delivered Ixo-vec at 3E10 and 1E11 vg/eye (human equivalent dose (HED) 6E10 and 2E11 vg/eye) doses in a GLP compliant 3 month-long study in Cynomolgus monkeys (Figure 1)
- Detection of Ixo-vec vgs and mRNA expression was accomplished using BaseScope, an in-situ hybridization technology

Figure 1. GLP-Compliant NHP Study Established Well-Tolerated Doses of Ixo-vec, which Express Clinically Meaningful Levels of Ocular Aflibercept

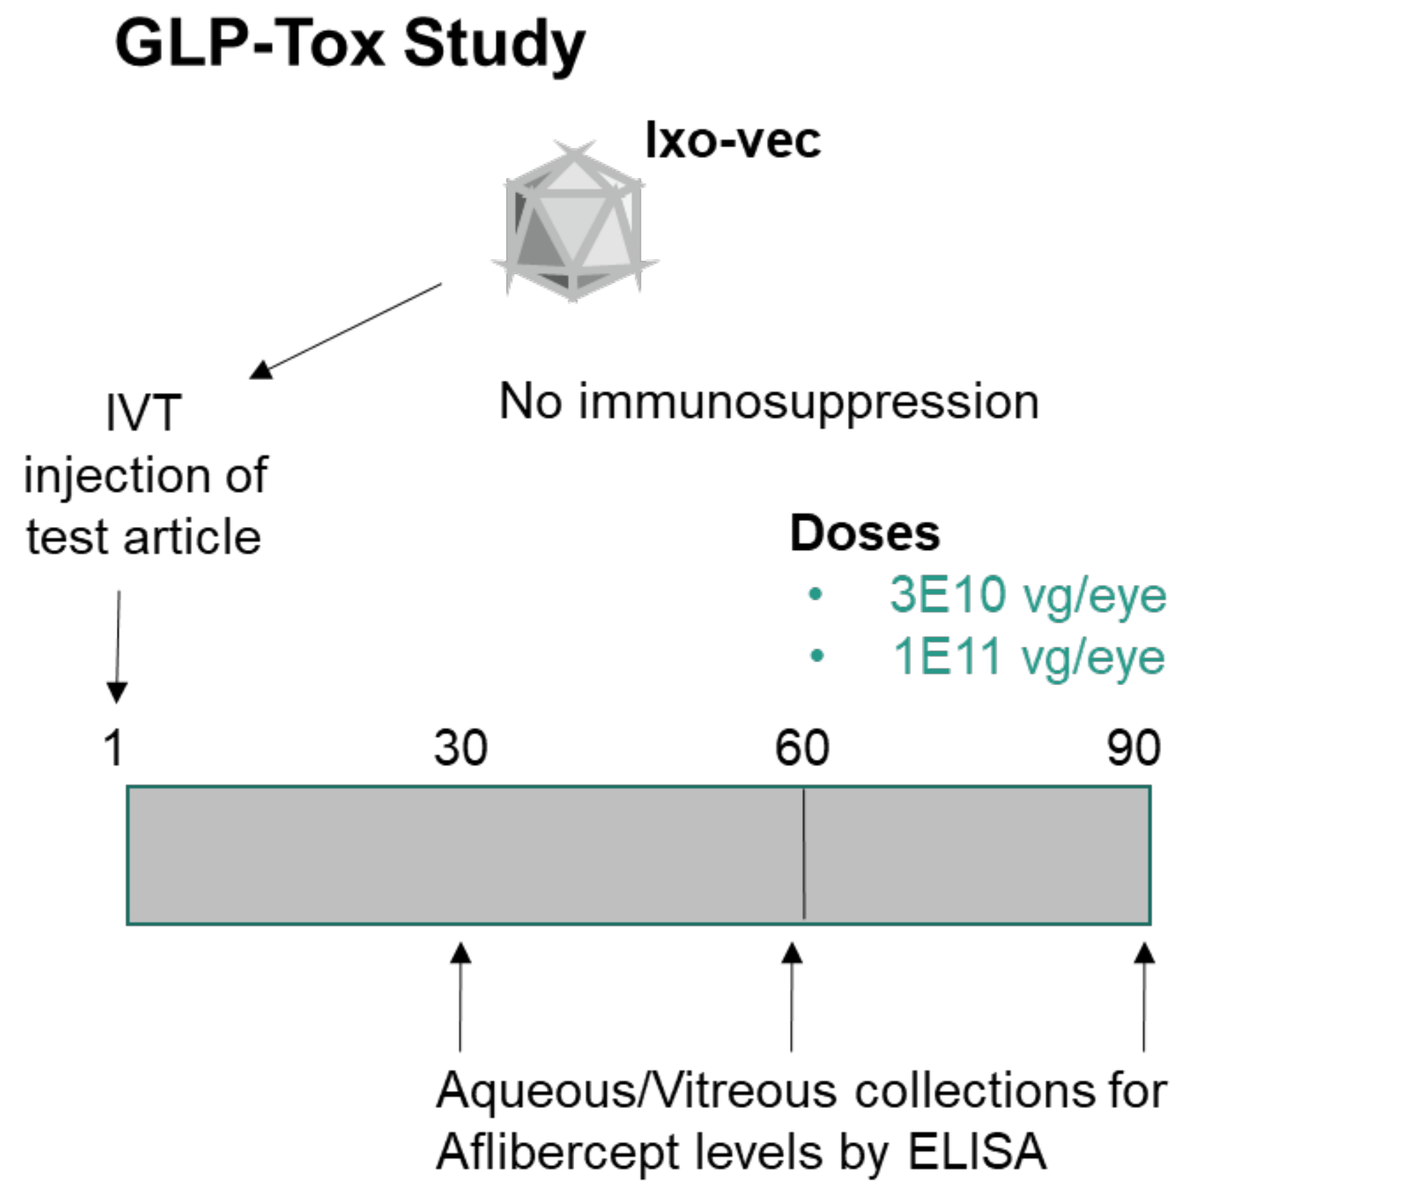
GLP-Tox Study
Ixo-vec
IVT
injection of
test article
No immunosuppression
Doses
3E10 vg/eye
1E11 vg/eye
1
30
60
90
Aqueous/Vitreous collections for
Aflibercept levels by ELISA

- No adverse systemic clinical signs observed
- Observations limited to non-adverse slight to mild dose dependent ocular inflammation characterized by pigment and vitreous cells
- No abnormalities in the anterior segment or lens
- IOP (intraocular pressure) within normal range
- Well-tolerated doses in the study express Aflibercept at levels comparable to those observed in human trial (Phase 1, OPTIC)
- Microscopic findings: minimal mononuclear cell infiltrates of minor severity considered non-adverse
- **No Observed Adverse Effect Level (NOAEL) identified at 1E11 vg/eye**

Ocular Safety Assessments
- Ophthalmic exams
- Tonometry (intraocular pressure)
- Optical Coherence Tomography (OCT)
- Electroretinography (ERG)

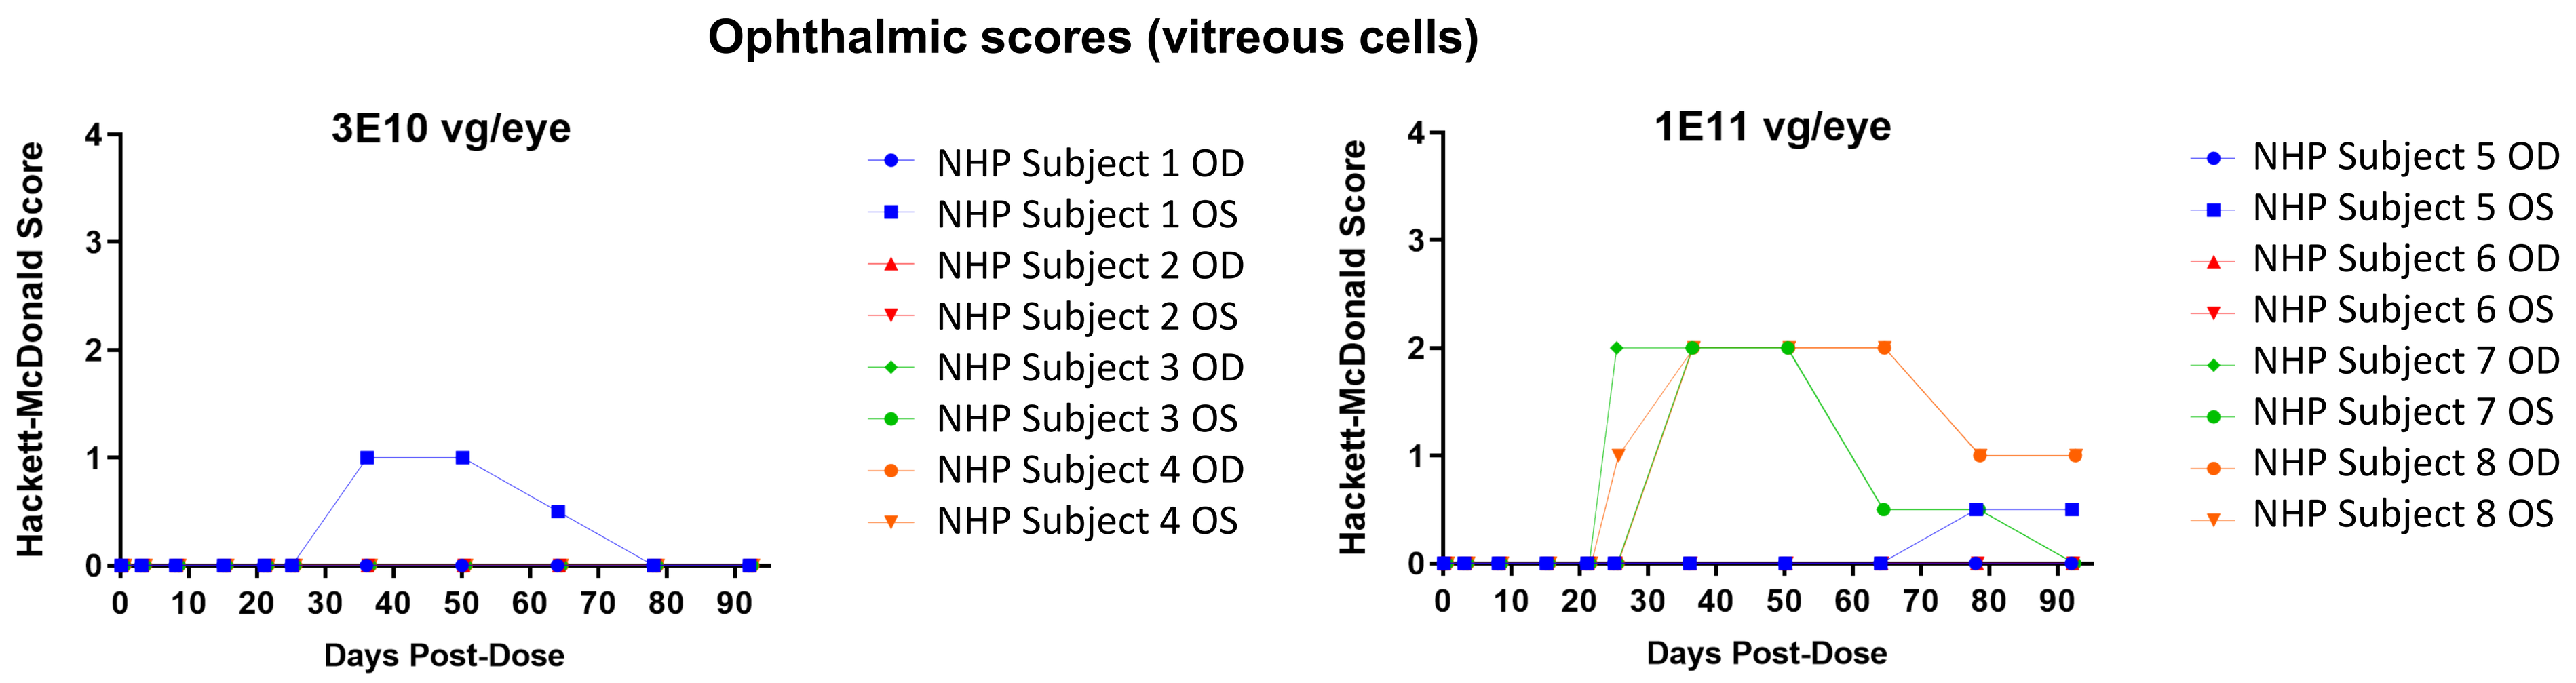
Ophthalmic scores (vitreous cells)
3E10 vg/eye
1E11 vg/eye
Hackett-McDonald Score
Days Post-Dose
NHP Subject 1 OD
NHP Subject 1 OS
NHP Subject 2 OD
NHP Subject 2 OS
NHP Subject 3 OD
NHP Subject 3 OS
NHP Subject 4 OD
NHP Subject 4 OS
NHP Subject 5 OD
NHP Subject 5 OS
NHP Subject 6 OD
NHP Subject 6 OS
NHP Subject 7 OD
NHP Subject 7 OS
NHP Subject 8 OD
NHP Subject 8 OS

METHODS

- Tissues were fixed in 10% neutral buffered formalin for 24 hours and embedded in paraffin
- Ixo-vec vector genome DNA was detected using a probe designed against the engineered promoter
- Aflibercept mRNA was detected using a 1-ZZ paired probe flanking the intron in 5′ UTR in the expression cassette. The probe generates a signal after the 5′ UTR intron has been spliced, which allows for the precise detection of transgene mRNA without vector DNA interference

RESULTS

Complete ocular vector genome coverage of IVT administered Ixo-vec DNA:

- Evaluation of intraocular biodistribution of Ixo-vec vgs in treated eyes, 3E10 vg/eye and 1E11 vg/eye (HED 6E10 vg/eye and 2E11 vg/eye) revealed widespread vg presence in posterior and anterior ocular tissues
- Retinal transduction was restricted to the macula and far-periphery in retina, where the inner limiting membrane (ILM) is the thinnest. Microscopic evaluation revealed vector transduction of retinal cells in the retinal ganglion cell (RGC) layer, inner nuclear cell layer (INL), and photoreceptor (PRL) layer (Figure 2)
- In the mid-periphery, where the ILM is the thickest, vgs were bound to the ILM and not found within the cells of the deeper layers of the retina (Figure 2)
- Wide-spread pattern of distribution of vgs suggests the role of intraocular fluid dynamics and drainage of humor. (Dörsam, 2019)

Ocular distribution of IVT-delivered Ixo-vec Aflibercept mRNA:

- Evaluation of intraocular biodistribution of Ixo-vec mRNA in treated eyes, 3E10 vg/eye and 1E11 vg/eye (HED 6E10 vg/eye and 2E11 vg/eye) revealed Aflibercept mRNA expression in anterior and posterior tissues with most prominent expression in the macula and far-periphery of the retina (Figure 3)
- Localization of Aflibercept mRNA expression in retina is influenced by ILM barrier
- Aflibercept mRNA expression was identified in retinal ganglion and inner-nuclear layer cells of the macula and far-periphery of the retina, and non-pigmented epithelium of anterior tissues (Figure 3)

Figure 2. Ubiquitous Ocular Distribution of IVT Administered Ixo-vec DNA

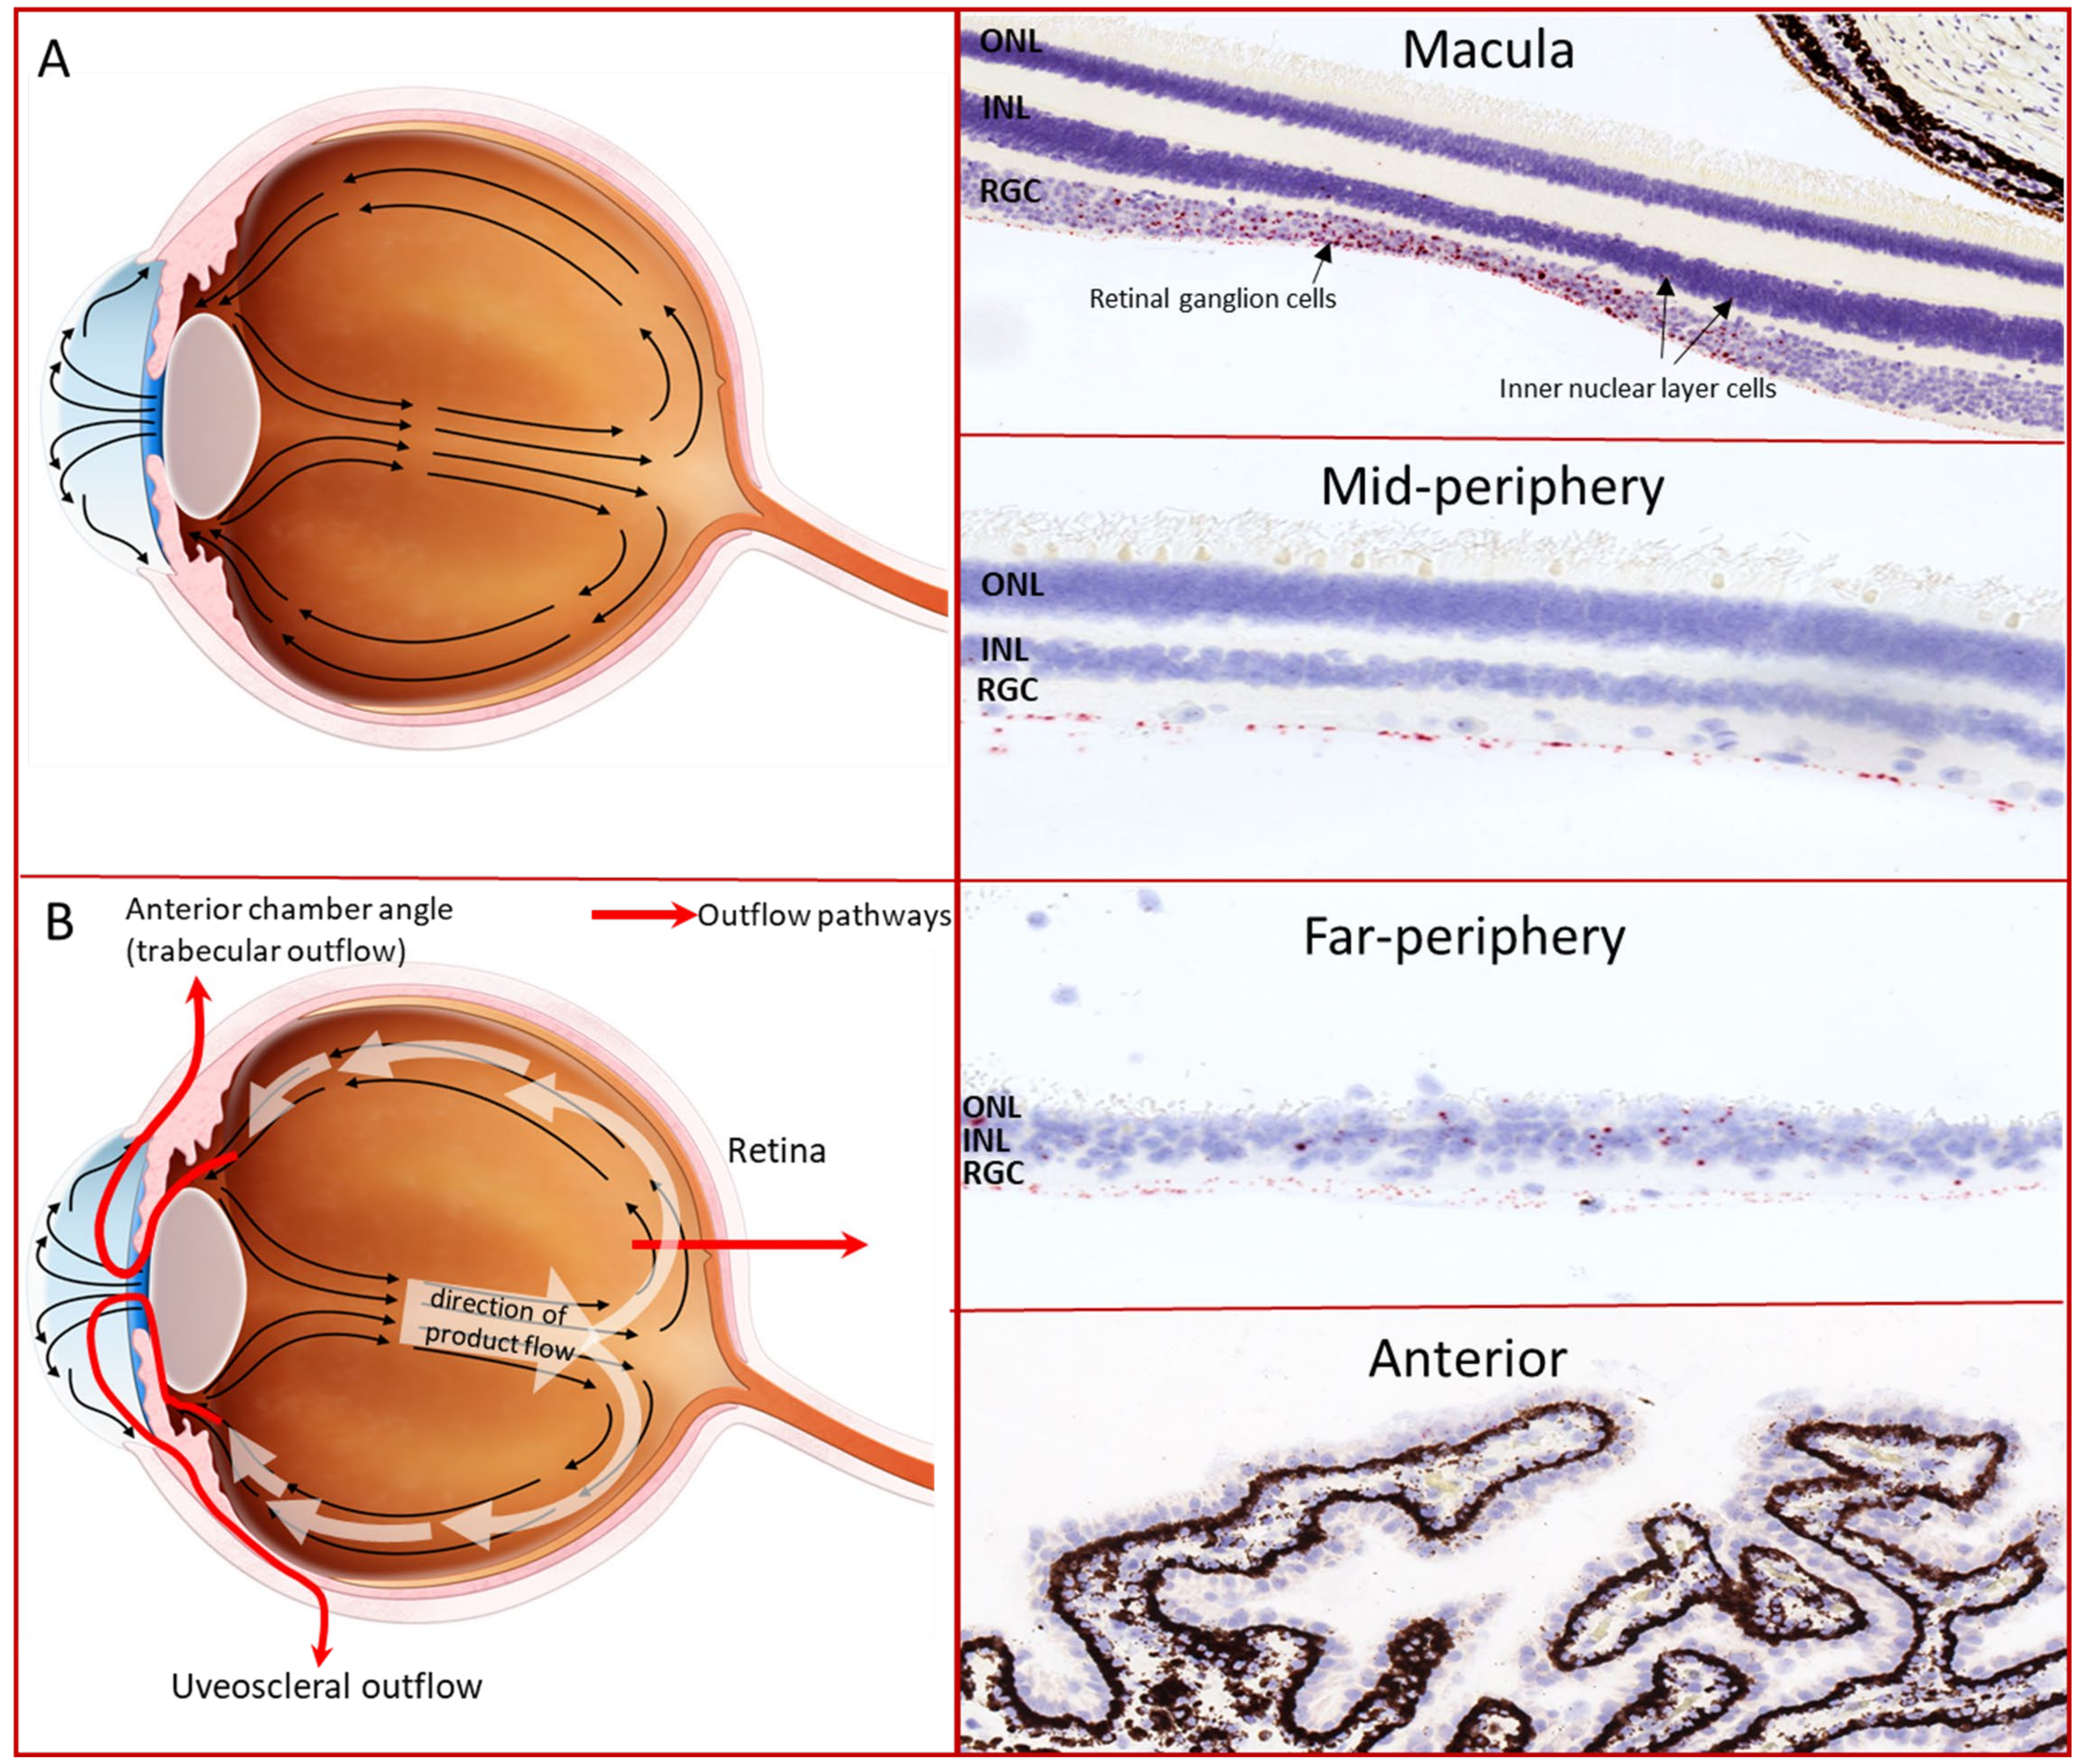
A
B
Anterior chamber angle
(trabecular outflow)
Outflow pathways
Retina
direction of
product flow
Uveoscleral outflow
Macula
ONL
INL
RGC
Retinal ganglion cells
Inner nuclear layer cells
Mid-periphery
Far-periphery
Anterior

ISH detected vector DNA in NHP eye treated with IVT-delivered Ixo-vec at 3E10 vg/eye dose (HED 6E10 vg/eye dose). Ixo-vec DNA biodistribution in 1E11 vg/eye (HED 2E11 vg/eye dose) dose mirrors results observed in 3E10 vg/eye dose. A) Modeling of intraocular fluid dynamics influenced by aqueous humor production (black arrows) B) Intraocular fluid flow-dependent distribution of IVT-delivered ocular gene therapy product (white arrows). Drainage of aqueous humor through anterior chamber angle, uveoscleral outflow, and retinal pathways. *RGC: retinal ganglion cell layer. INL: inner nuclear cell layer. ONL: outer nuclear layer. Red arrows: passage of aqueous humor through drainage facilities. Black arrows: Flow of aqueous humor into anterior and posterior eye. Red: vector DNA. Purple: nuclei.*

CONCLUSIONS

- Intraocular fluid convection together with anterior and posterior fluid outflow influences ubiquitous distribution of vgs from IVT-delivered ocular gene therapy products
- Localization of Ixo-vec mRNA expression indicates influence of ILM barrier.
- Expression from retinal and nonretinal tissues can contribute to pharmacologically active ocular levels of Aflibercept
- Well-tolerated doses and per cell evaluation of Ixo-vec biodistribution and expression is supported by transcriptome results *presented by Dr. Julian Ramos ASGCT May 18, 2023 – 2PM PST Room 411*

Figure 3. Ixo-vec Drives Expression of Aflibercept in Macula and Far-periphery

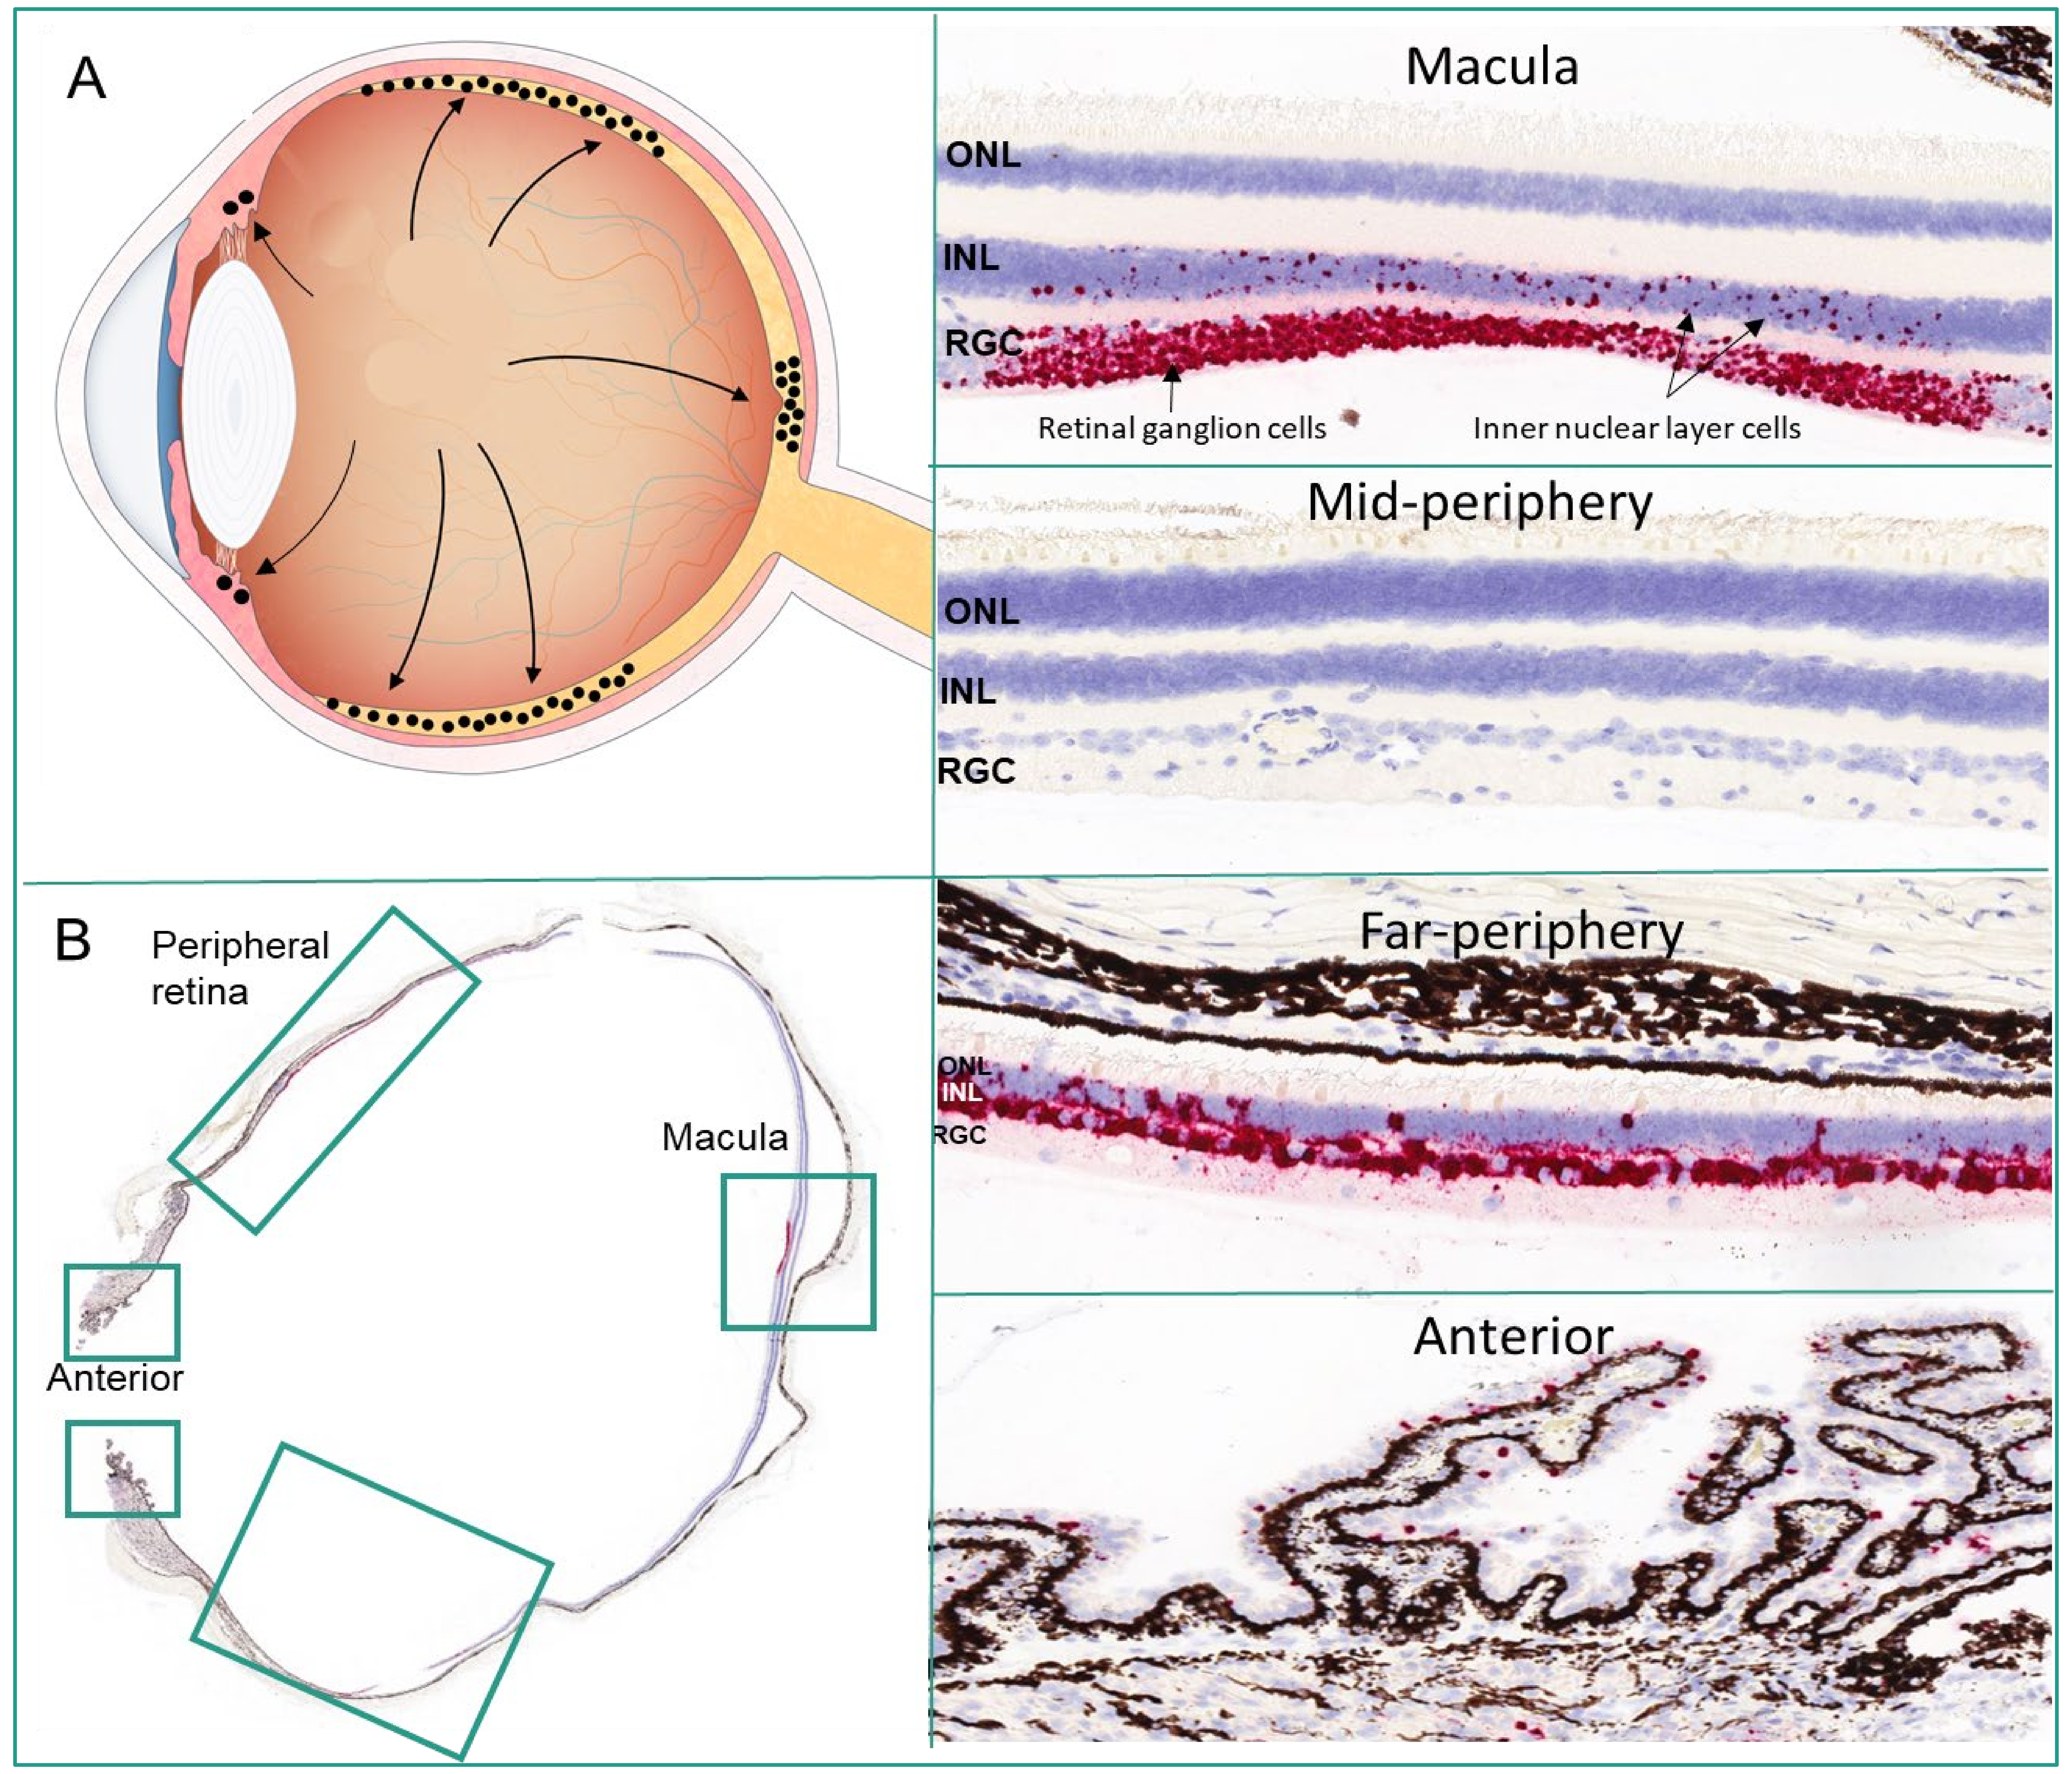
A
B
Peripheral
retina
Macula
Anterior
ONL
INL
RGC
Retinal ganglion cells
Inner nuclear layer cells
Mid-periphery
Far-periphery

ISH detected Aflibercept mRNA expression in NHP eye treated with IVT-delivered Ixo-vec at 3E10 vg/eye dose (HED 6E10 vg/eye dose). Ixo-vec expression in 1E11 vg/eye dose (HED 2E11 vg/eye dose) mirrors results observed in 3E10 vg/eye dose. A) Localization of mRNA expression in the eye. *RGC: retinal ganglion cell layer. INL: inner nuclear cell layer. ONL: outer nuclear layer. Red: Aflibercept mRNA expression. Purple: nuclei.*

Acknowledgements: ***Advanced Cell Diagnostics & Gardenia Gonzalez Gil with Living Pixels Biomedical Illustrations***

Disclosures: ***The authors are employees at Adverum Biotechnologies, Inc., and hold shares at the company***

References:
1. Dörsam S, Olkhovskiy V, Friedmann E. Modeling and simulation of the aqueous humor flow in the human eye. Proc Appl Math Mech. 2019;19(1):e201900462. doi:10.1002/PAMM.201900462.