Functional and molecular evaluation of intravitreal (IVT) AAV gene therapy vectors for the treatment of Geographic Atrophy (GA)

Gustavo de Alencastro, Anna Zaslavsky, Stephanie Lussier, Kelly Michiko Hanna, Julio Nieves, Jenny Vo, Jie Ma, Cameron K. Baker, Ali Aghajanirefah, Ruslan N. Grishanin, Brigit E. Riley

ADVERUM

Contact: Gustavo de Alencastro - gdealencastro@adverum.com

Background and reasons for using IVT AAV injection for treatment of GA

- Dry age-related macular degeneration (Dry AMD) with geographic atrophy (GA) is a highly prevalent disease, characterized by retinal pigment epithelium (RPE) and photoreceptor death leading to vision loss, which affects the quality of life in the aging population worldwide.
- Several of the complement components have shown strong genome wide association (GWAS) to Dry AMD, and data from approved products support inhibition of the complement pathway as a therapeutic strategy for Dry AMD.
- Best in class therapeutics may need to address the runaway complement amplification loop <u>on both sides</u> of the outer blood/retinal barrier (Figure 1).

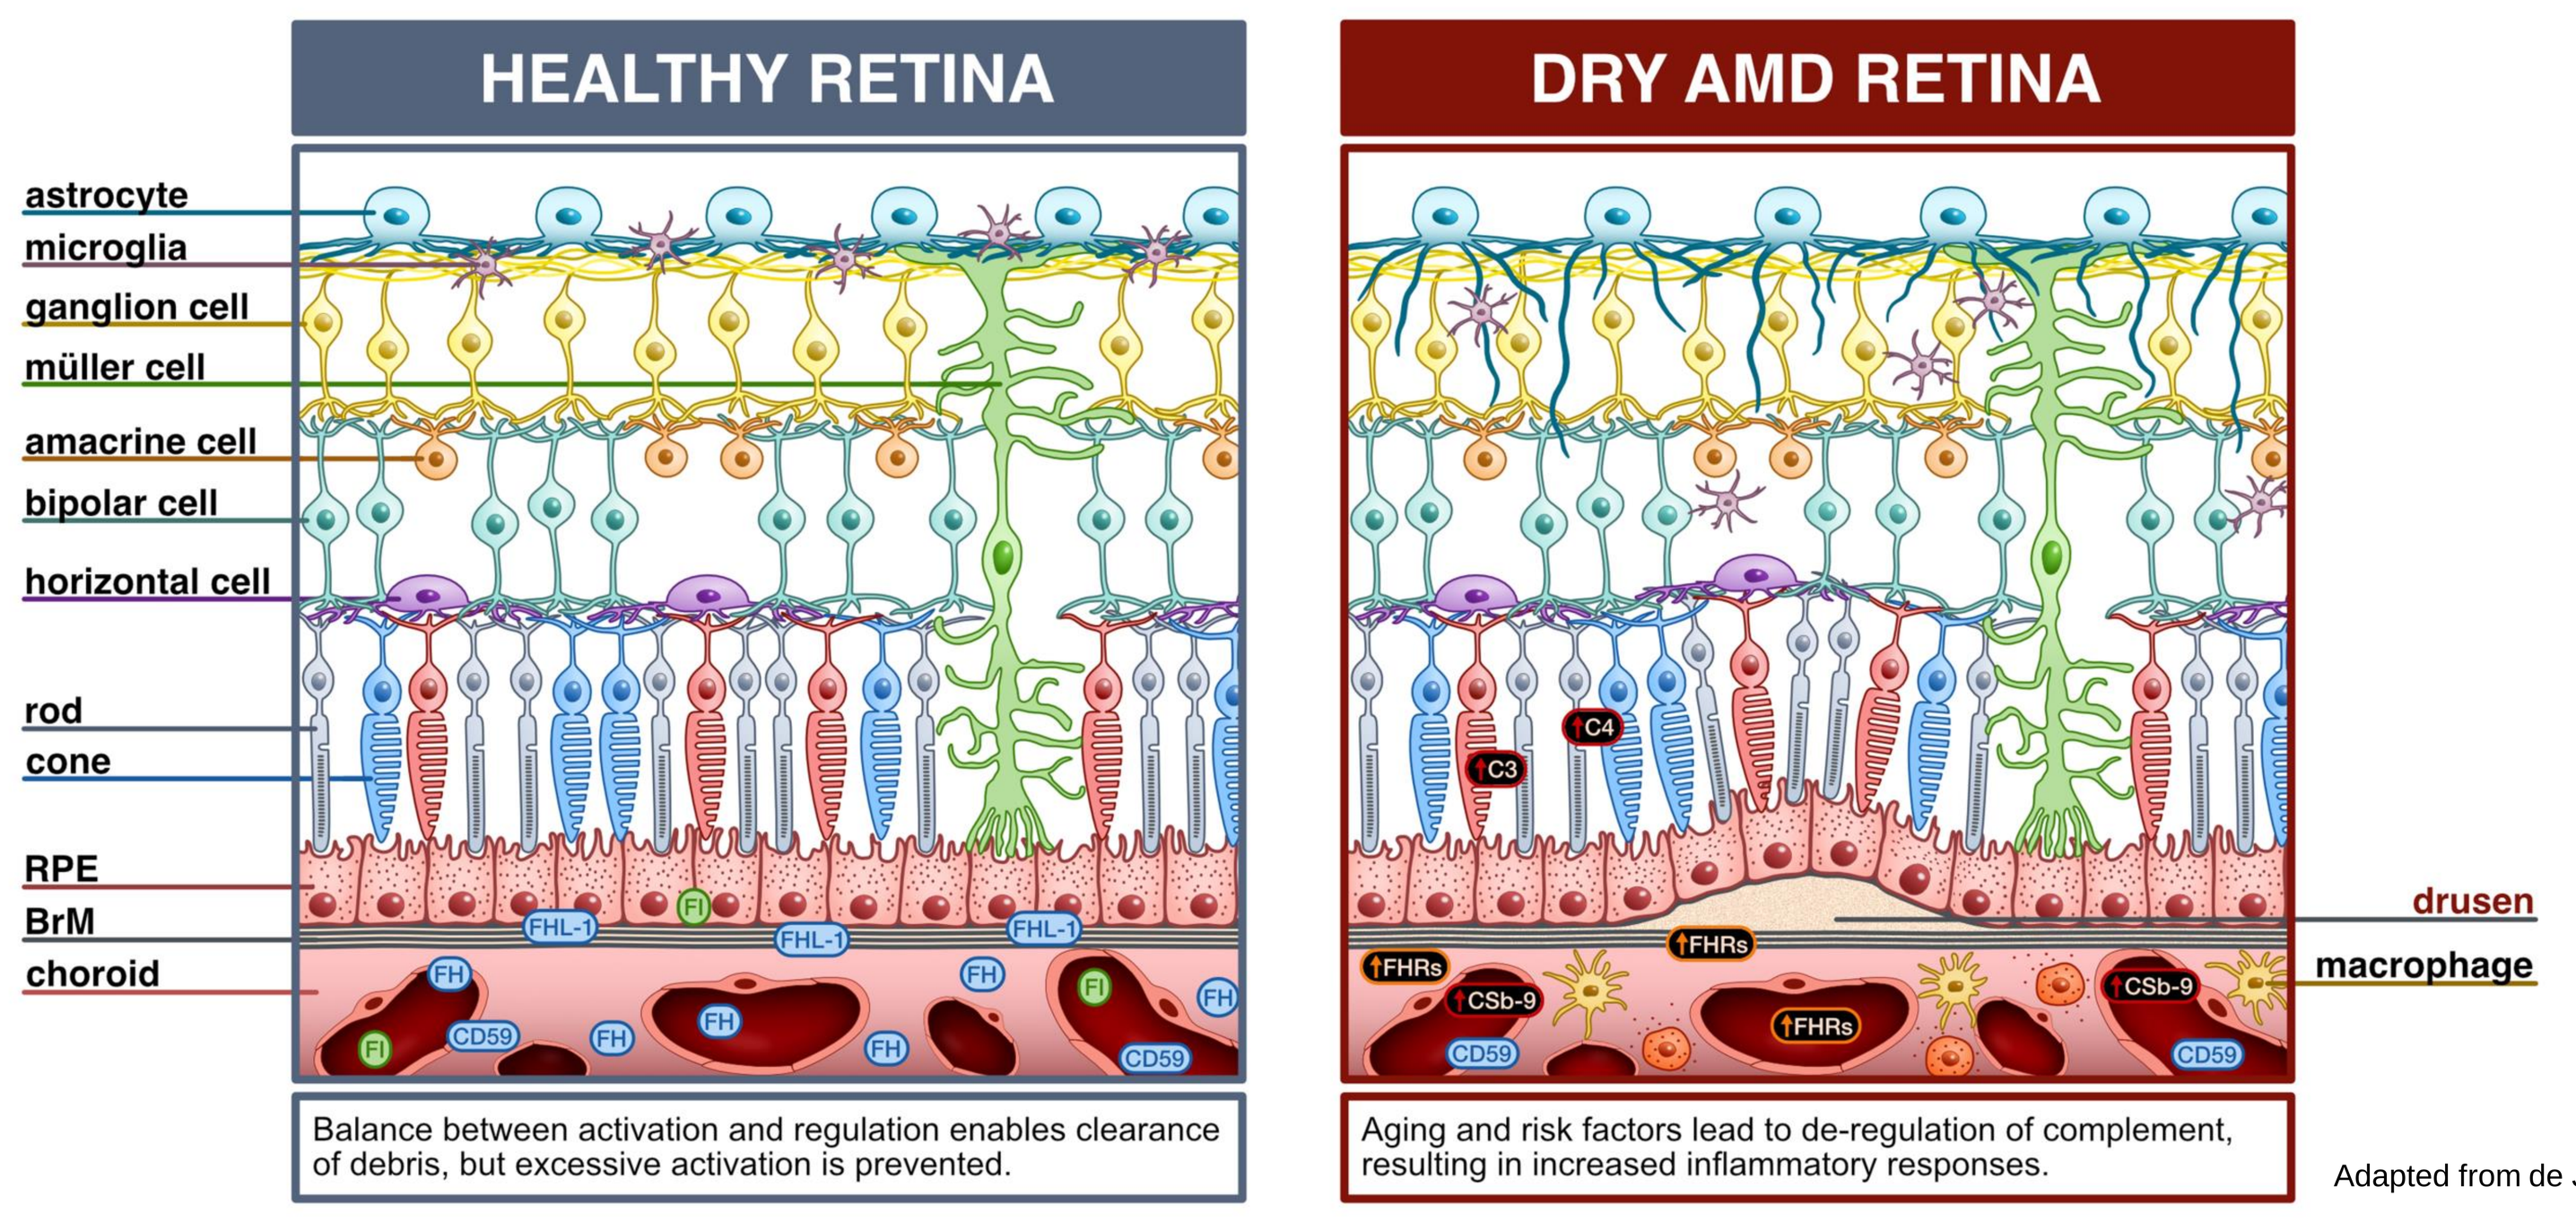
HEALTHY RETINA
DRY AMD RETINA
astrocyte
microglia
ganglion cell
müller cell
amacrine cell
bipolar cell
horizontal cell
rod
cone
RPE
BrM
choroid
drusen
macrophage
FHL-1
FI
FH
CD59
C3
C4
FHRs
C5b-9
Balance between activation and regulation enables clearance of debris, but excessive activation is prevented.
Aging and risk factors lead to de-regulation of complement, resulting in increased inflammatory responses.

Adapted from de Jong *et al.*, 2023

Figure 1. Complement regulation in the retina. In the healthy retina presence of fluid phase and surface-bound regulators like complement Factor I (FI) and Factor H (FH), and CD59 prevent aberrant complement activation (left panel). Impairment of these regulators or excess presence of the de-regulating FH related (FHR) proteins negatively affect this regulatory balance, and excess deposition of C5b-9, C3, and C4 leads to local tissue damage (right panel).

- Complement Factor I (CFI) is a rate-limiting enzyme in the complement cascade. Variants in the CFI gene are linked to a higher risk of developing Dry AMD. **A single administration of an AAV vector via routine in-office IVT procedure (Figure 2A) is an ideal therapy for highly prevalent ocular disorders such as Dry AMD.**

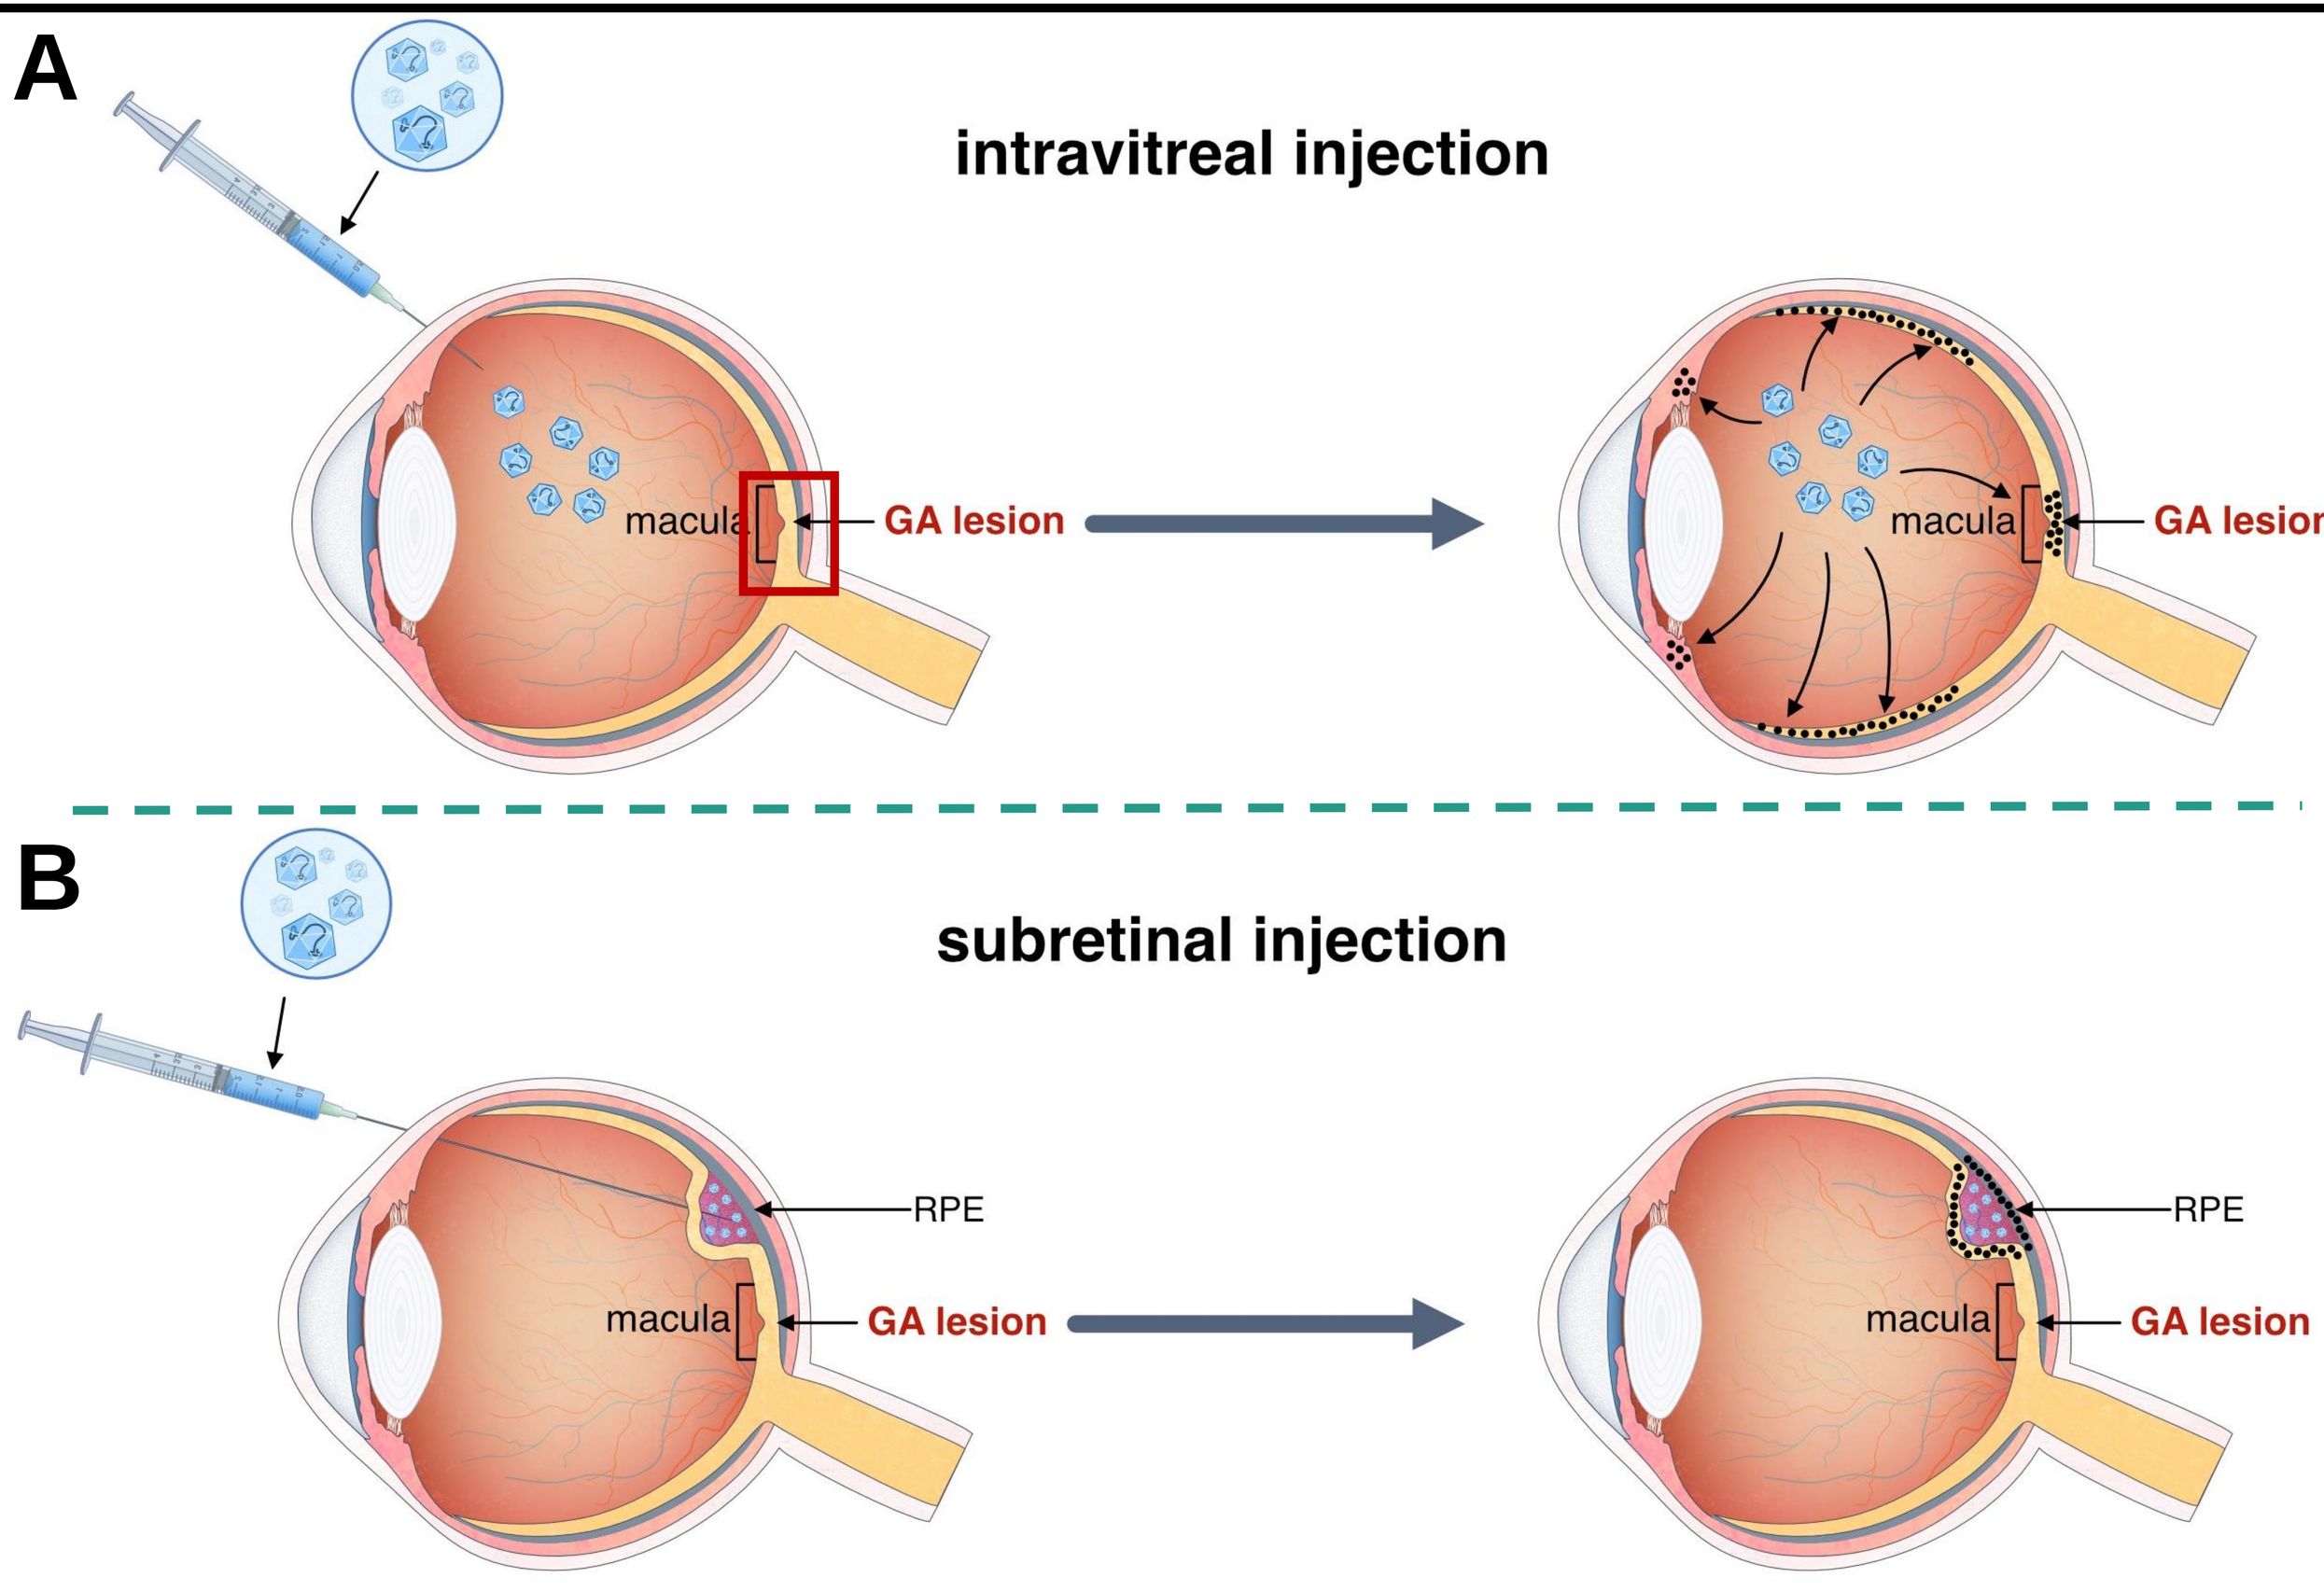
A
intravitreal injection
macula
GA lesion
B
subretinal injection
RPE

Limitations of subretinal injection for Dry AMD therapy.

- Subretinal injection can induce mechanical damage, loss of retinal function and reactive gliosis due to the injection procedure.
- AAV expression after subretinal injection is primarily from photoreceptor cells (and some RPE) around the bleb.
- Correct processing of CFI is dependent on the presence of intracellular furin (CFI is first produced as a Pro-CFI). Transducing a limited number of cells around the bleb with subretinal injection, furin could be exhausted – can potentially impair processing of endogenous proteins.
- Gene therapy products generated after subretinal injection of the AAV will have to be transported into the vitreous humor, then across the retina layers, to reach GA foveal lesions/choroid RPE – efficiency is untested.

Figure 2. Comparison of the two main routes of administration (RoA) of AAV vectors for ocular disorders. **(A)** After intravitreal (IVT) injection using retinotropic AAV vectors, such as AAV2.7m8 and AAV-LSV1, broad retinal transduction is achieved, especially in the macula - the major region impacted in patients with GA (indicated by the red box). **(B)** On the other hand, subretinal injection of AAVs will result in a localized transduction (around the bleb), which limits overall AAV expression in the ocular cavity and macular expression (site of the GA lesion).

Methods and non-human primate (NHP) study design

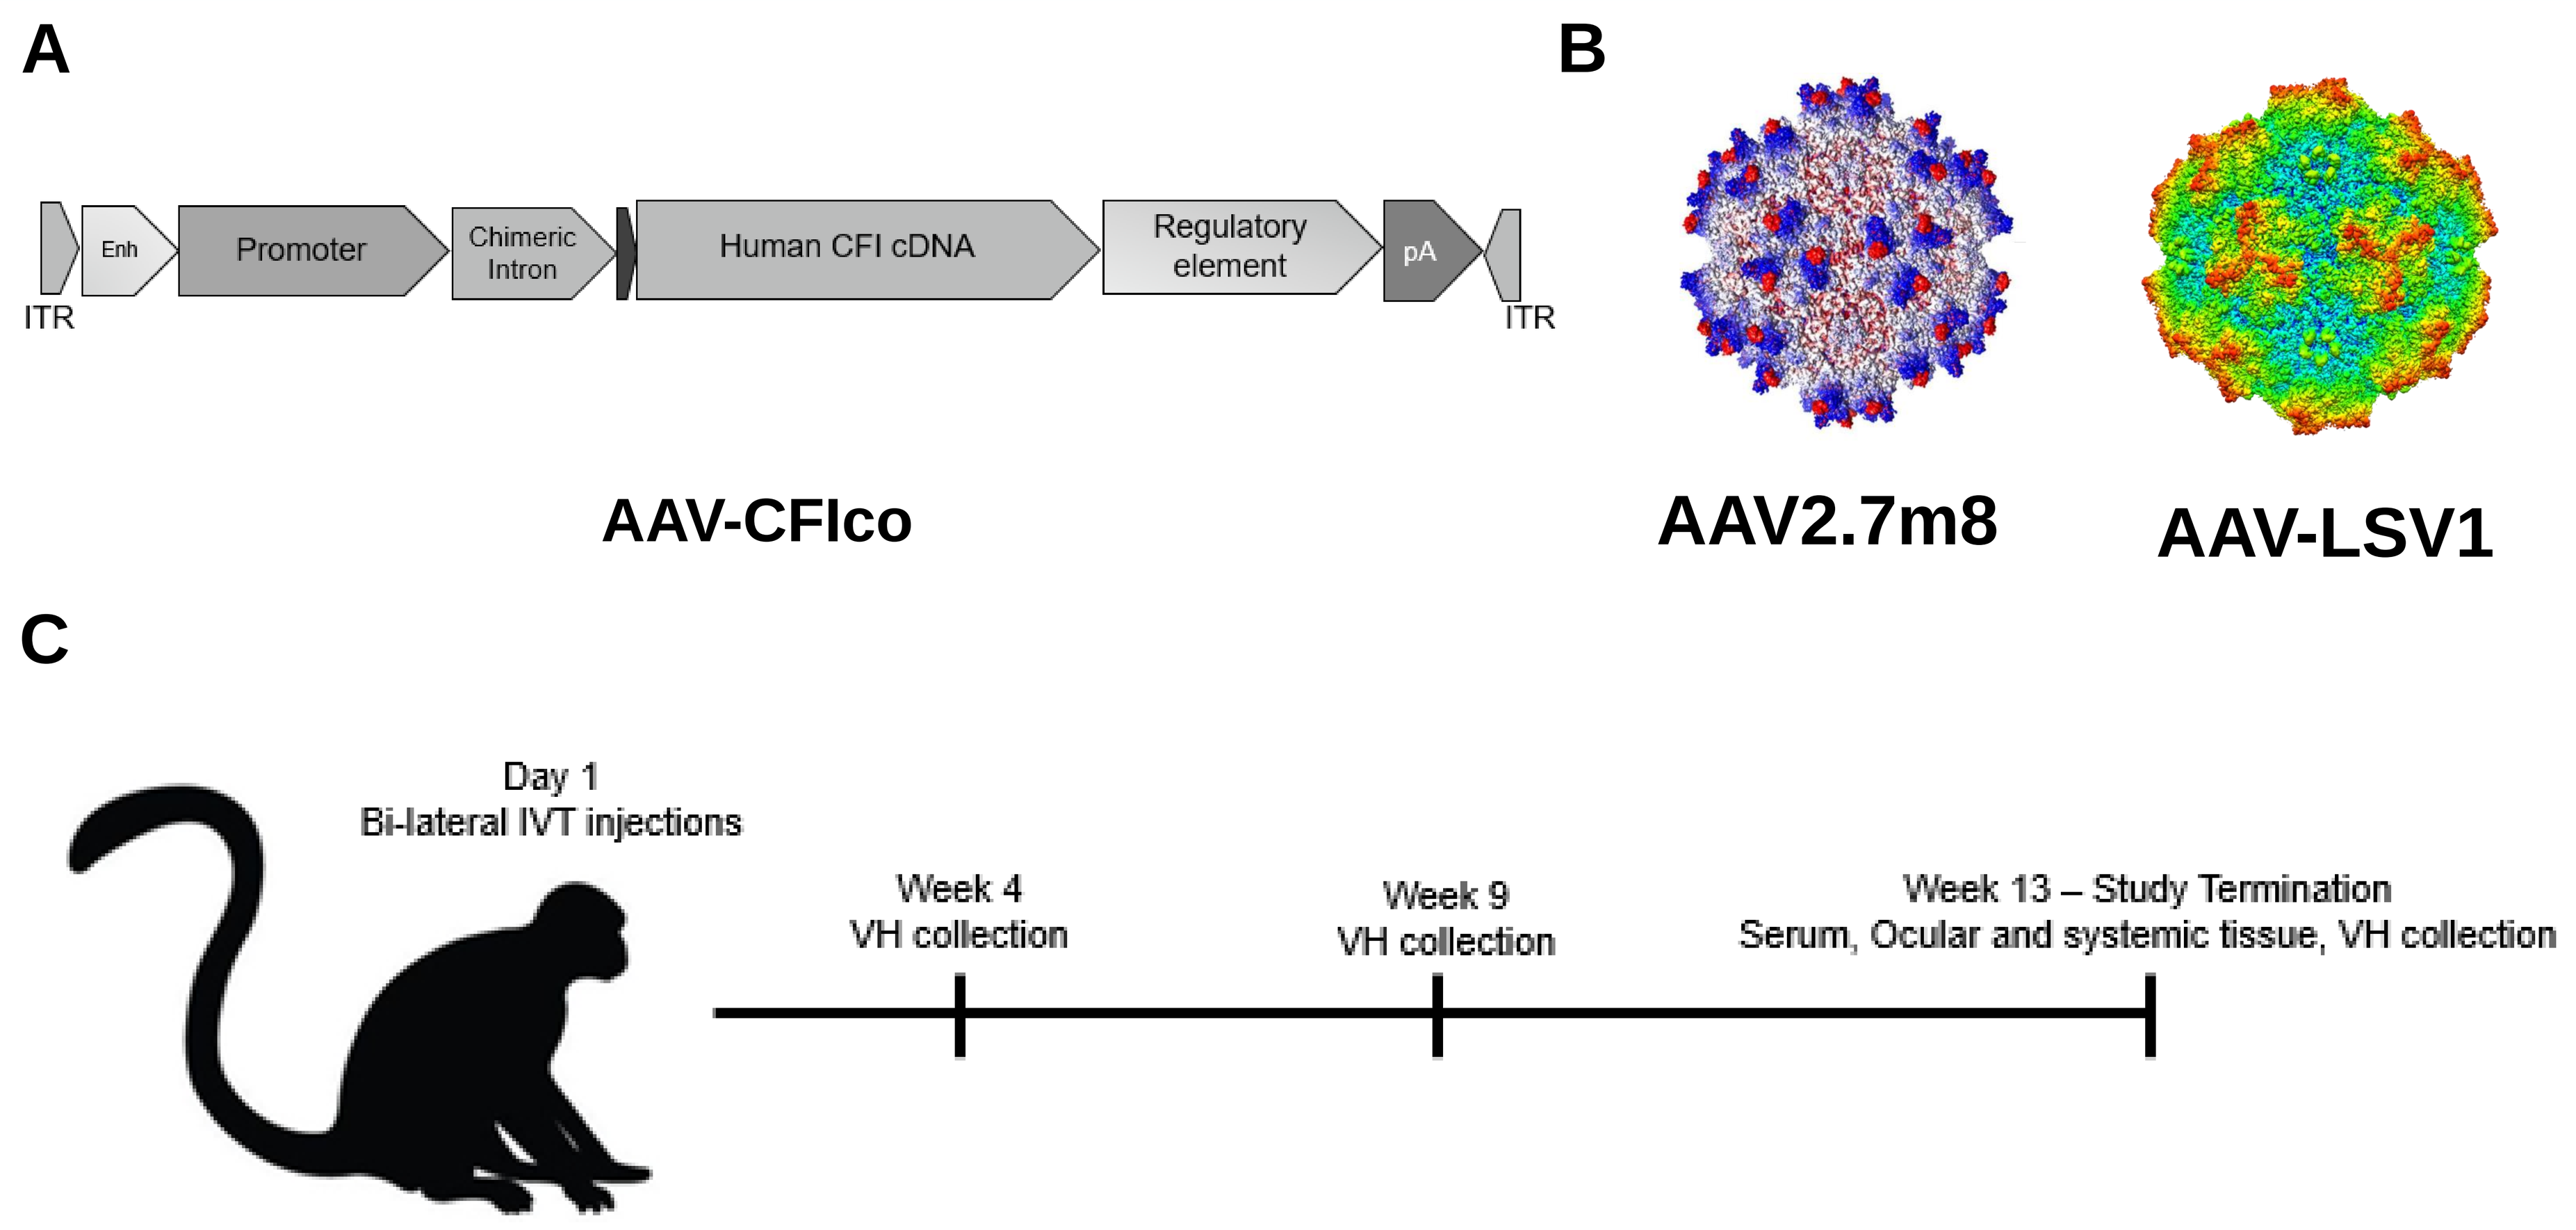
A
Enh
Promoter
Chimeric Intron
Human CFI cDNA
Regulatory element
pA
ITR
ITR
AAV-CFIco
B
AAV2.7m8
AAV-LSV1
C
Day 1
Bi-lateral IVT injections
Week 4
VH collection
Week 9
VH collection
Week 13 – Study Termination
Serum, Ocular and systemic tissue, VH collection

Table 1. Summary of NHP study design.

Group	Test Material	Serotype	Dose Level (vg/eye)	Human Equivalent Dose, HED (vg/eye)	Dose Volume (mL/eye)	Number of NHP subjects
1	Vehicle	N/A	0	0	0.05	3
2	AAV-CFIco	AAV2.7m8	3E10	6E10	0.05	3
3	AAV-CFIco	AAV2.7m8	1E11	2E11	0.05	3
4	AAV-CFIco	AAV-LSV1	3E10	6E10	0.05	3
5	AAV-CFIco	AAV-LSV1	1E11	2E11	0.05	3

Figure 3. A codon-optimized human CFI sequence (CFIco) was cloned into an AAV vector backbone and packaged using Adverum's proprietary engineered AAV2.7m8 and AAV-LSV1 capsids for IVT delivery.
(A) The human CFIco cDNA was inserted in a strong expression cassette flanked by AAV2 inverted terminal repeats (ITRs). **(B)** Cryogenic electron microscopy images of both retinotropic AAV chimeric capsids (AAV2.7m8 and AAV-LSV1) for IVT injection. AAV2.7m8 was identified in screens across species from mouse, canine and non-human primate (NHP), and contains a peptide insertion. AAV-LSV1 was identified from a NHP screen and contains a loop swap variant. These capsid modifications allow the AAV vectors to bypass the inner limiting membrane (ILM) to efficiently transduce and deliver transgenes to target retinal cells. **(C)** Scheme of the NHP study performed to evaluate tolerability and human CFI expression in the vitreous humor (VH) and ocular tissues after IVT injection of AAV-CFIco vector packaged into both the AAV2.7m8 and AAV-LSV1 serotypes. NHP subjects were negative for neutralizing antibodies for AAV2.7m8 (Groups 2 and 3) and for AAV-LSV1 (Groups 4 and 5). Enh: enhancer; pA: polyA.

Results

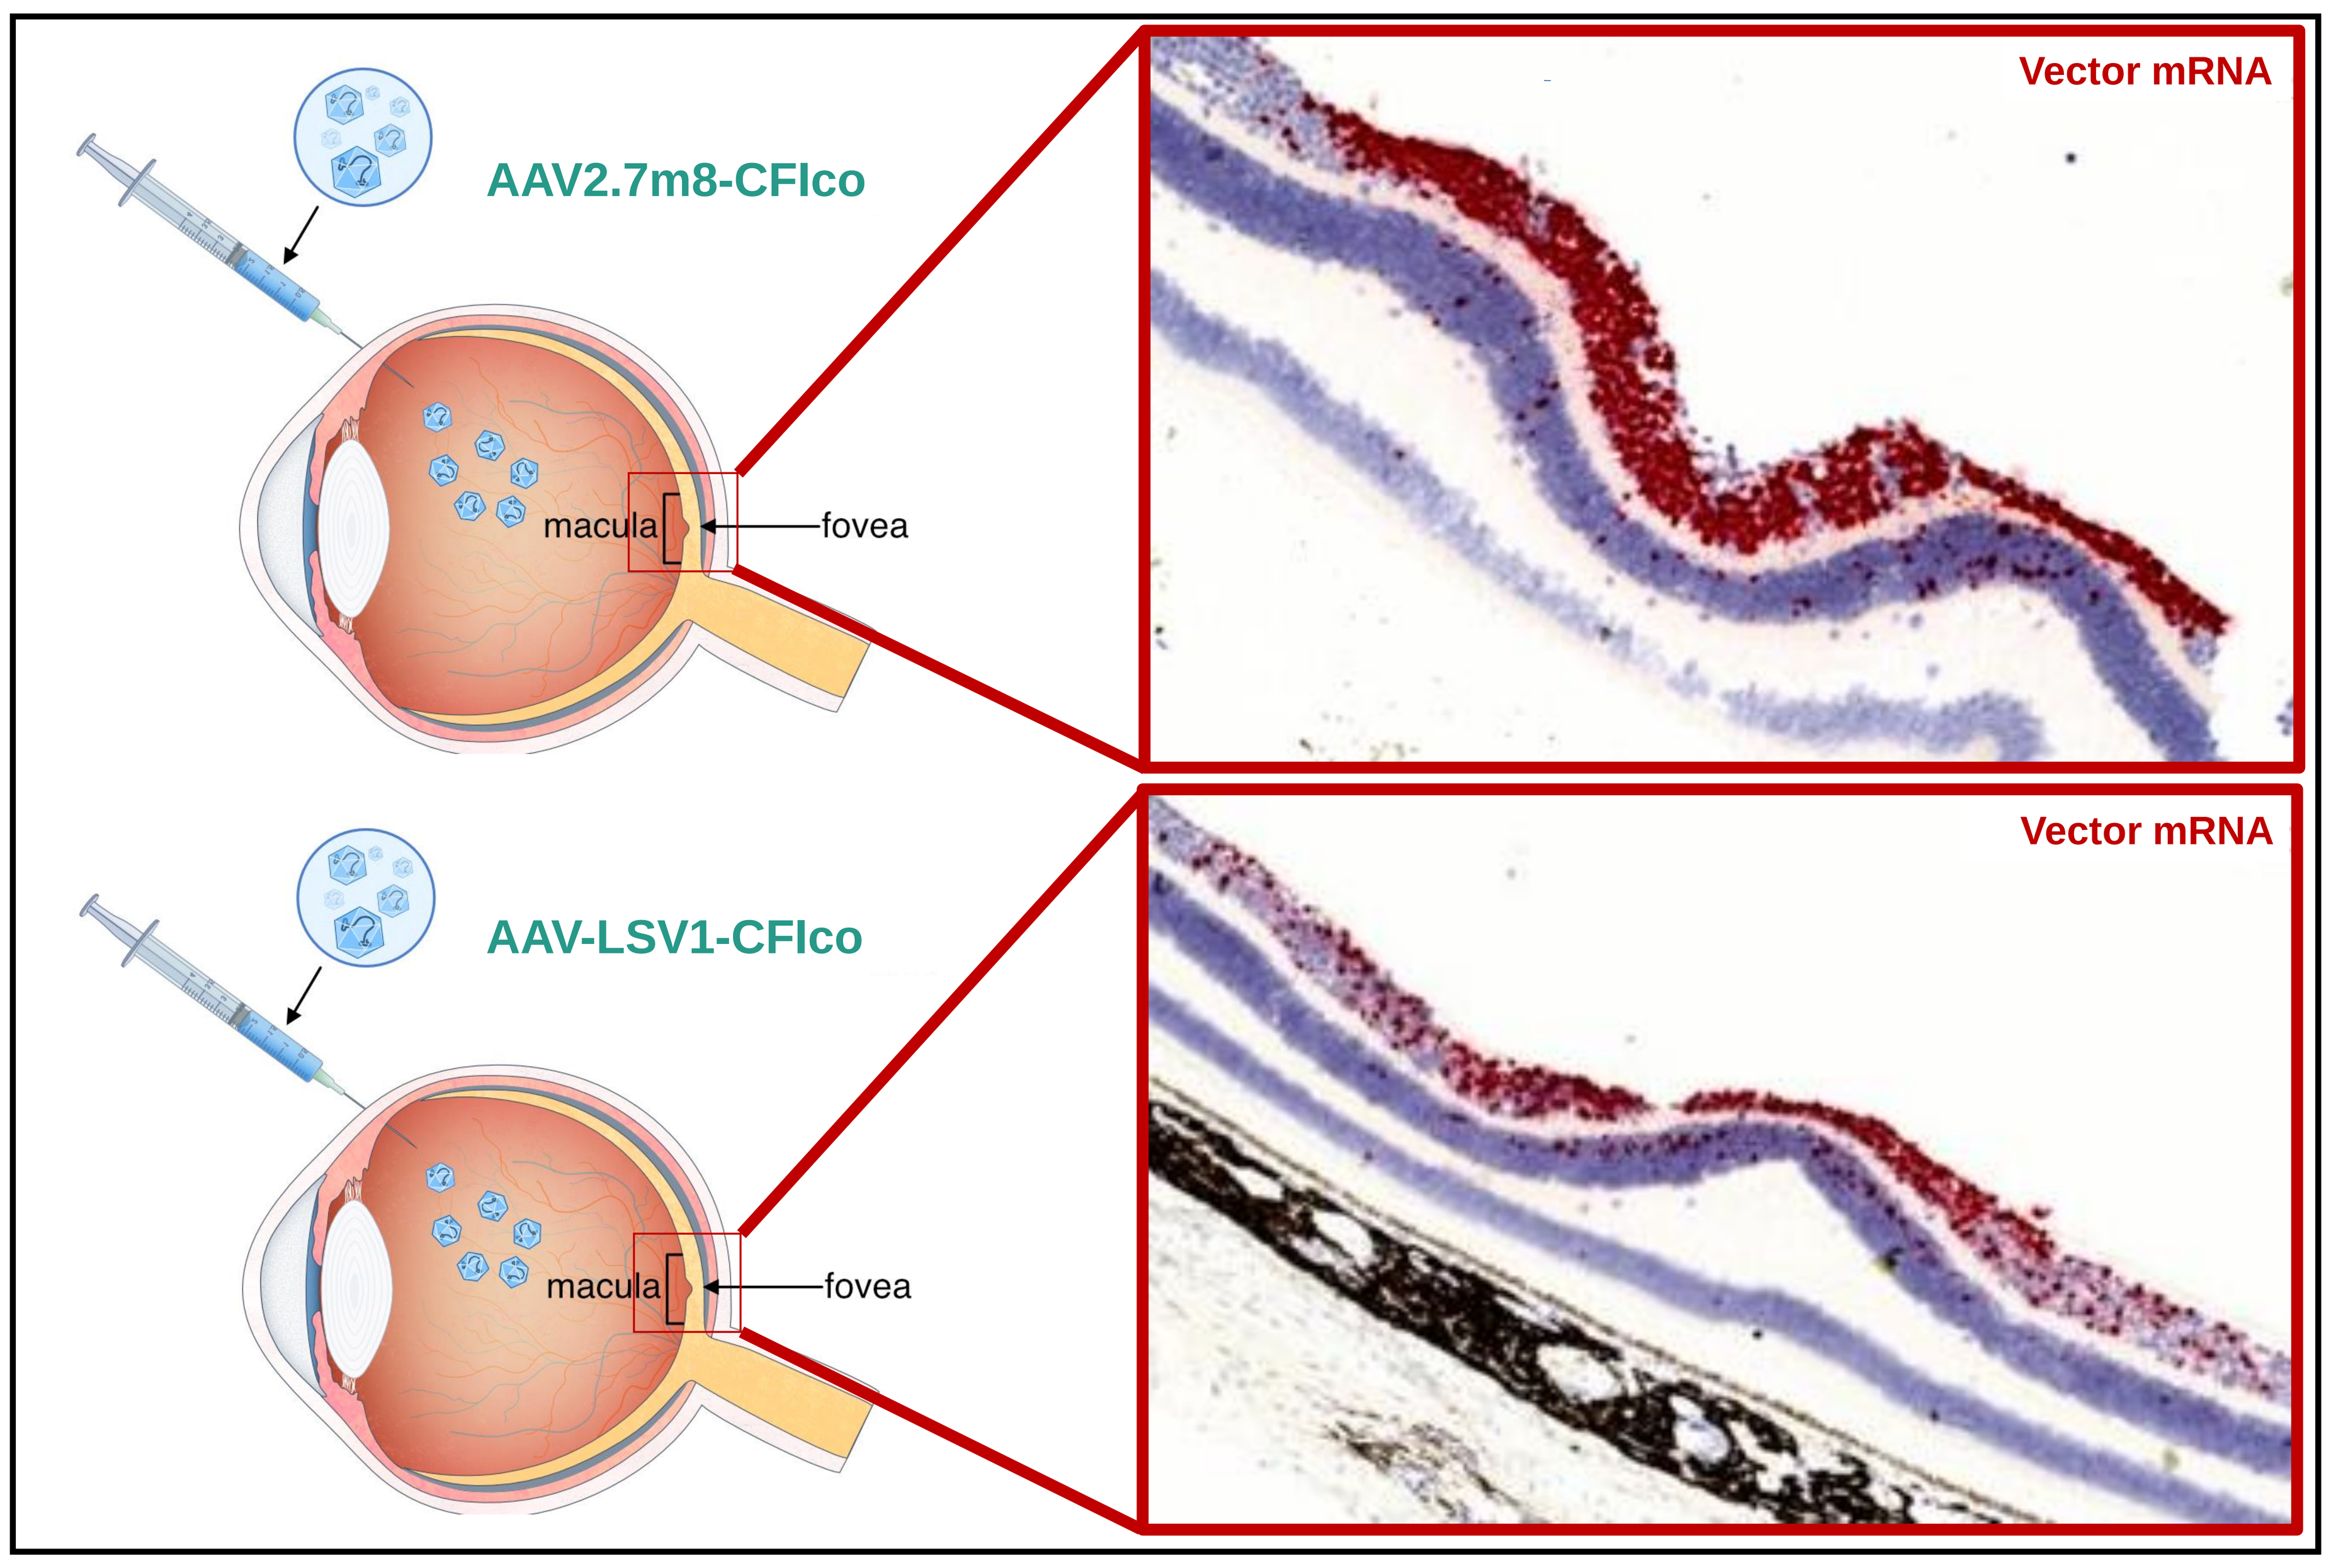
AAV2.7m8-CFIco
macula
fovea
Vector mRNA
AAV-LSV1-CFIco
macula
fovea
Vector mRNA

Figure 4. IVT administered AAV2.7m8 and AAV-LSV1 demonstrate robust macula transduction - the major region impact in patients with GA. Vector mRNA detection in NHP retina sections by in situ hybridization using probes specific for mRNA. For both AAV2.7m8-CFIco and AAV-LSV1-CFIco, mRNA is mainly produced in the macula (by retinal ganglion cells and photoreceptors), retina (far-periphery) and ciliary process. Human CFI protein secreted by transduced cells is detected in the retina/choroid tissue, the areas affected in GA, as shown in Table 2.

Table 2. Presence of hCFI protein in different ocular compartments and anti-drug antibody (ADA) in serum of NHP subjects injected with AAV-CFIco packaged into both AAV2.7m8 and AAV-LSV1 capsids. Data obtained using samples collected at study termination. For CFI (ng/mL in VH and ng/g in tissues): **-**: BLOQ, **+**: 1-100, **++**: 101-600, **+++**: >601 (Luminex assay). ADA formation was anticipated given the amino acid differences between the human and NHP CFI proteins. For ADA (ECL values): **-**: <199, **+**: 200-10000, **++**: 10001-100000, **+++**: 100001-1000000.

Group/Serotype/Dose	NHP subject	hCFI in VH	hCFI in Retina (including Choroid)	hCFI in ICB	ADA in Serum
1 Vehicle	1	-	-	-	-
	2	-	-	-	-
	3	-	-	-	-
2 AAV2.7m8 3E10 vg/eye	4	+	++	++	++
	5	+++	+++	+++	-
	6	+	++	++	++
3 AAV2.7m8 1E11 vg/eye	7	-	+	-	+++
	8	++	+++	++	++
	9	+++	+++	+++	-
4 AAV-LSV1 3E10 vg/eye	10	+++	+++	++	+
	11	+	+	+	+++
	12	++	++	++	+
5 AAV-LSV1 1E11 vg/eye	13	++	+++	+++	+
	14	-	+	-	+++
	15	-	-	-	+++

Conclusions

- AAV-CFIco packaged into both AAV2.7m8 and AAV-LSV1 capsids provided robust levels of ocular human CFI.
- IVT administration of AAV2.7m8 and AAV-LSV1 demonstrated efficient transduction of the macula region – the area affected in GA patients.
- CFI expressed from AAVs seem to diffuse to all regions affected in GA – retina and choroid – potentially the only therapeutic capable of delivering an inhibitor of the complement system to these regions.
- AAV-CFIco with both AAV2.7m8 and AAV-LSV1 capsids was well tolerated by NHP subjects.
- The biology of IVT injected AAVs is remarkably distinct from those injected subretinally - different ocular regions, especially the macula, can be efficiently targeted by IVT injection using retinotropic AAVs, and more cells can be modified using a routine in-office procedure, ideal for highly prevalent ocular disorders such as Dry AMD.

Disclosures: *The authors are employees at Adverum Biotechnologies, Inc., and hold shares at the company.*

Acknowledgements: *Charles River Laboratory (CRL) ran the NHP study, performed ophthalmic evaluations and developed ADA assay. ACD bio performed RNAscope analysis. Robert Mckenna and Antonette D. Bennett from UFL for generating LSV1 Cryo-EM. Gardenia Gonzalez Gil for illustrations.*